iCELLis500: A versatile Manufacturing Platform for CGT and Oncolytic Virus Production

- Lessons Learned and Best Practices from Successful Programs -

Dr. Martin Pech[1], Sabrina Pelz[1&2], Cihan Sözen[2], Stefan Augustin[1], Thomas Rautenberg[1] , Elke Pommerening[1], Julia Janson[1], Frank Mahrla[1], Michael Kijas[1], Mathias Kahl[2]
[1]MSAT, IDT Biologika, [2]Development , IDT Biologika

1. Introduction

Over 100 years in biopharmaceutical manufacturing

End-to-End Support

Regulatory Compliance EMA, FDA, ANVISA approved

Global Recognition Repeated award honors over multiple years

IDT Biologika is a global biopharmaceutical **contract development and manufacturing organization** and part of SK Bioscience specializing in **cell culture** and microbial based proteins**, vaccines, gene & immune therapeutics,** biosimilars and other sterile injectables (Fig.1). Within this poster we present two phase III client projects. An AAV and an oncolytic virus (*Parvovirus*) project using the iCELLis500+ platform.

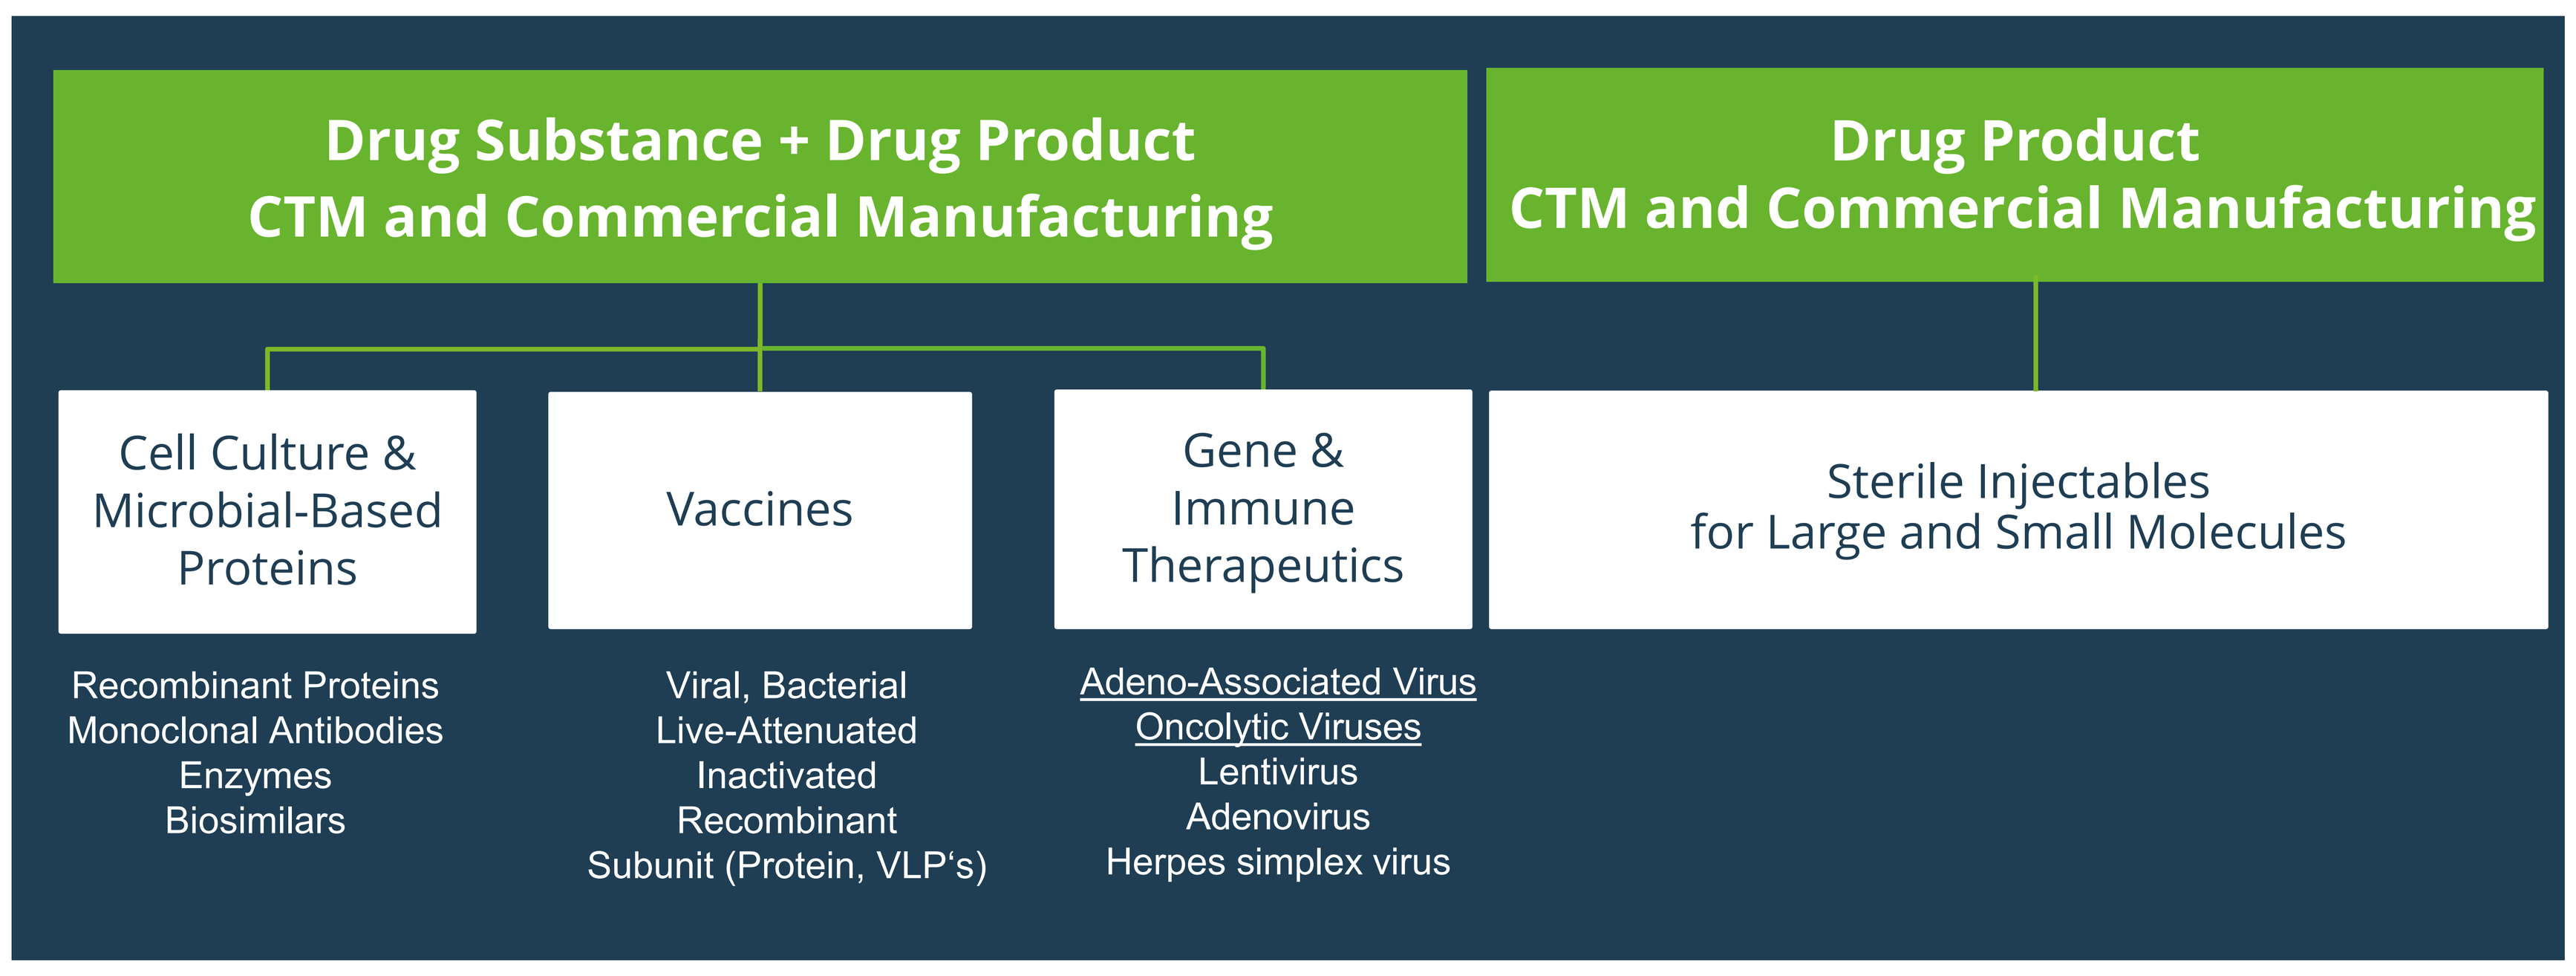
Drug Substance + Drug Product
CTM and Commercial Manufacturing
Drug Product
CTM and Commercial Manufacturing
Cell Culture & Microbial-Based Proteins
Vaccines
Gene & Immune Therapeutics
Sterile Injectables for Large and Small Molecules
Recombinant Proteins
Monoclonal Antibodies
Enzymes
Biosimilars
Viral, Bacterial
Live-Attenuated
Inactivated
Recombinant
Subunit (Protein, VLP's)
Adeno-Associated Virus
Oncolytic Viruses
Lentivirus
Adenovirus
Herpes simplex virus

Fig.1: IDT Biologikas key strength in clinical and commercial services in DS & DP manufacturing.

2. Available drug substance technologies

IDT Biologika has implemented a wide rang of scalable state of the art technologies in USP and DSP for end-to-end services. IDT provide all services beginning with process and analytical development, scale ups and ending in clinical trail or commercial manufacturing **(Fig.2 & Fig.3)** .

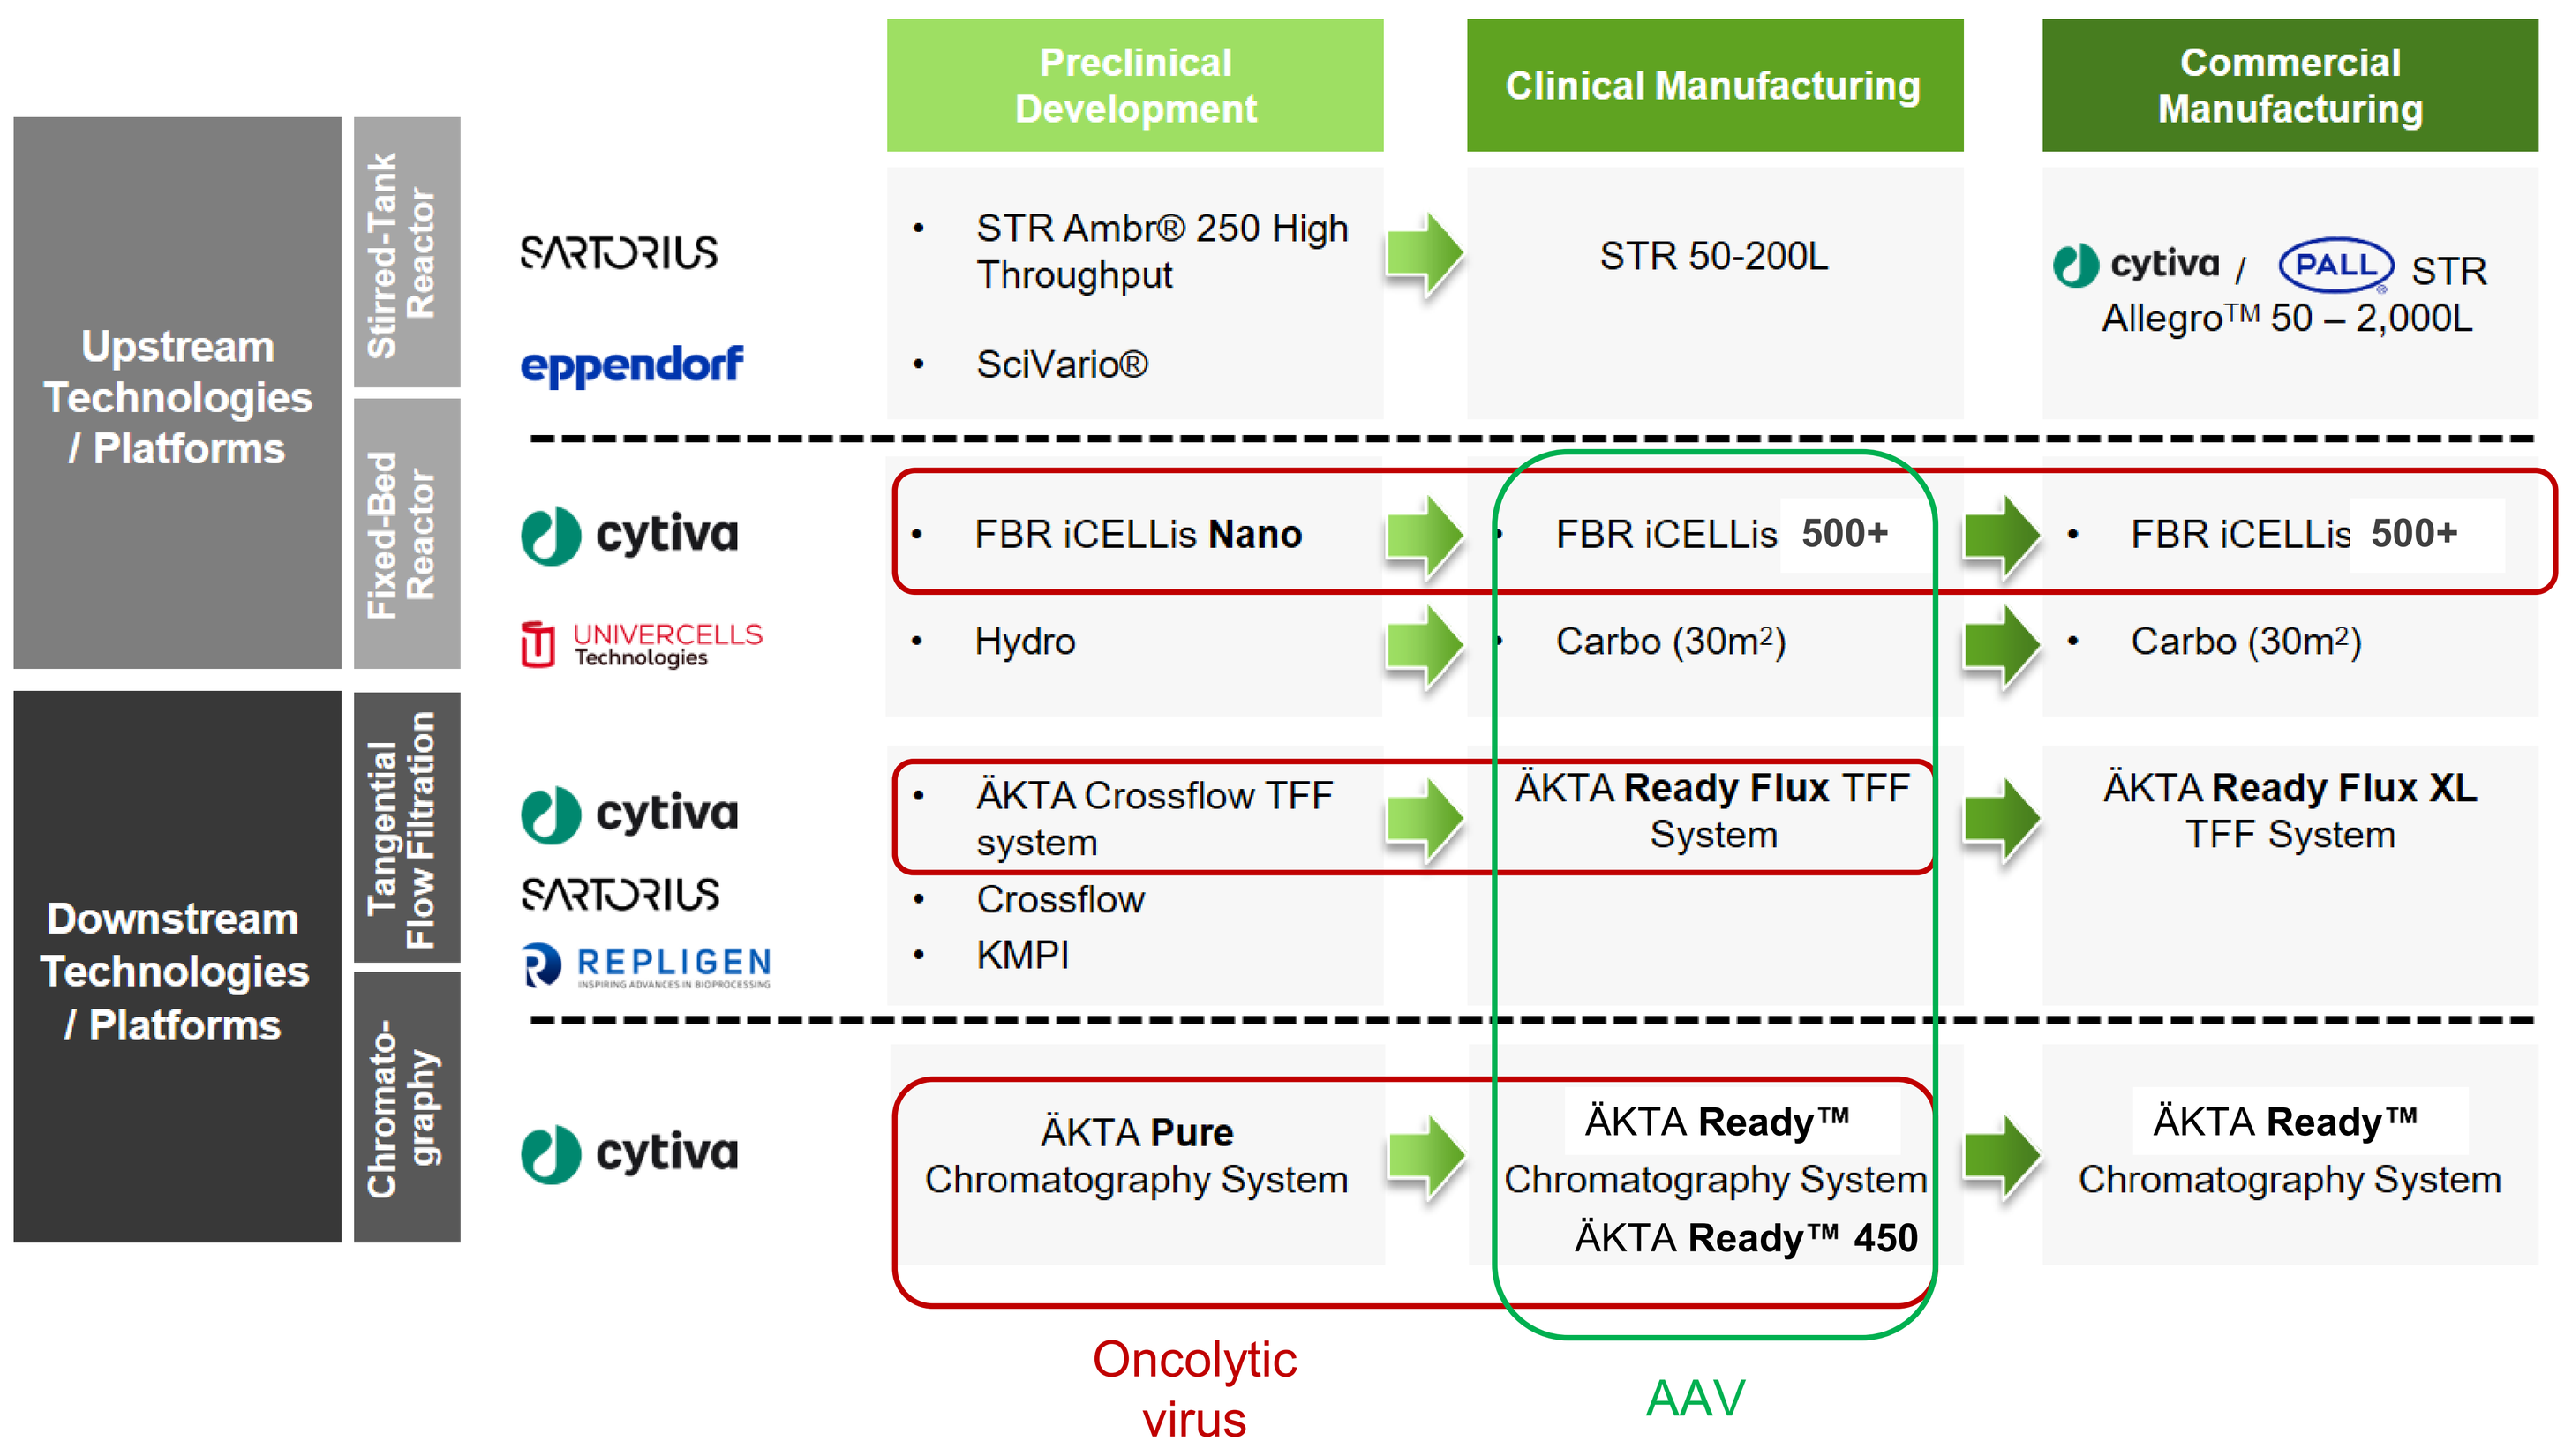
Preclinical Development
Clinical Manufacturing
Commercial Manufacturing
Upstream Technologies / Platforms
Stirred-Tank Reactor
Fixed-Bed Reactor
Downstream Technologies / Platforms
Tangential Flow Filtration
Chromato-graphy
STR Ambr® 250 High Throughput
SciVario®
STR 50-200L
cytiva / PALL STR Allegro™ 50 – 2,000L
FBR iCELLis Nano
FBR iCELLis 500+
FBR iCELLis 500+
Hydro
Carbo (30m²)
Carbo (30m²)
ÄKTA Crossflow TFF system
Crossflow
KMPI
ÄKTA Ready Flux TFF System
ÄKTA Ready Flux XL TFF System
ÄKTA Pure Chromatography System
ÄKTA Ready™ Chromatography System
ÄKTA Ready™ 450
ÄKTA Ready™ Chromatography System
Oncolytic virus
AAV

Fig.2: Overview of scalable USP and DSP technologies at IDT Biologika for process development and manufacturing.

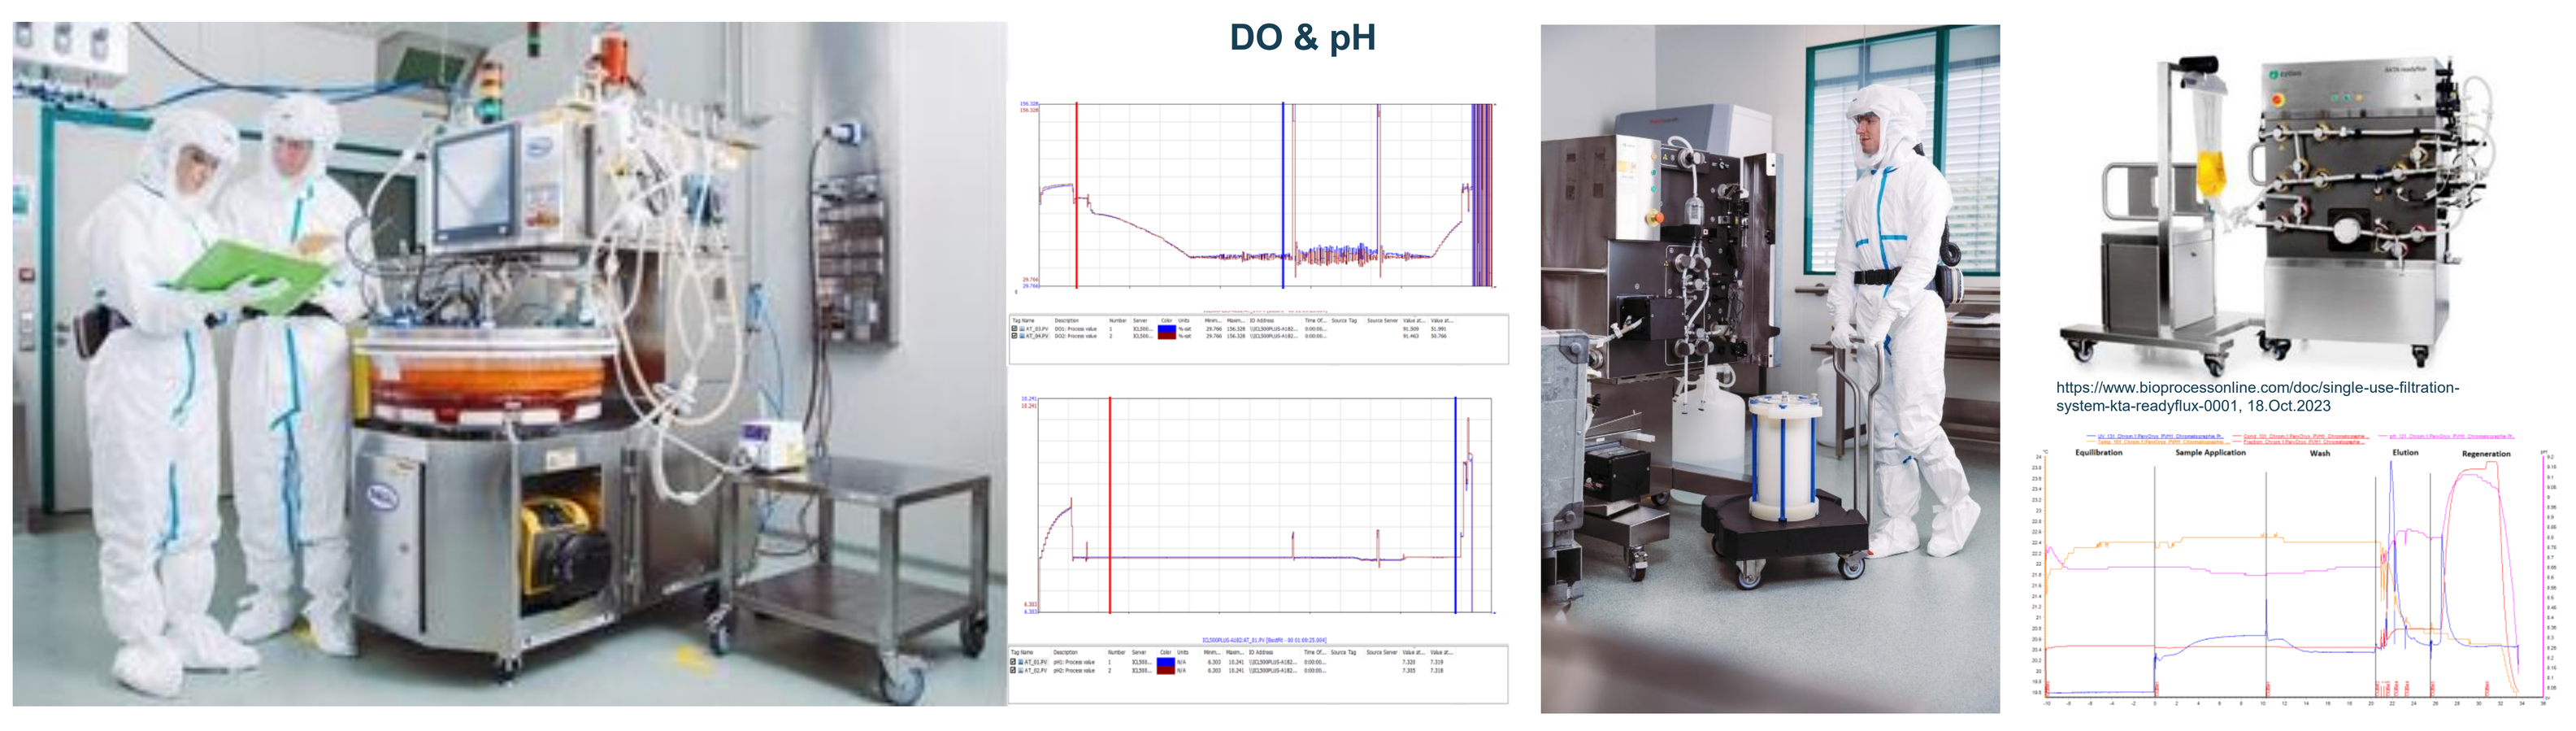
DO & pH
https://www.bioprocessonline.com/doc/single-use-filtration-system-kta-readyflux-0001, 18.Oct.2023

Fig.3: Insides in the manufacturing suites (BSL-3) and implemented technologies like the fixed bed bioreactor iCELLis 500+, chromatography system ÄKTA ready, tangential flow filtration (TFF) system ÄKTA readyflux and insights in the implemented software tools (Lucullus PIMS and Unicorn 7.11) for process automation and data collection.

3. IDT Biologika's process implementation road map

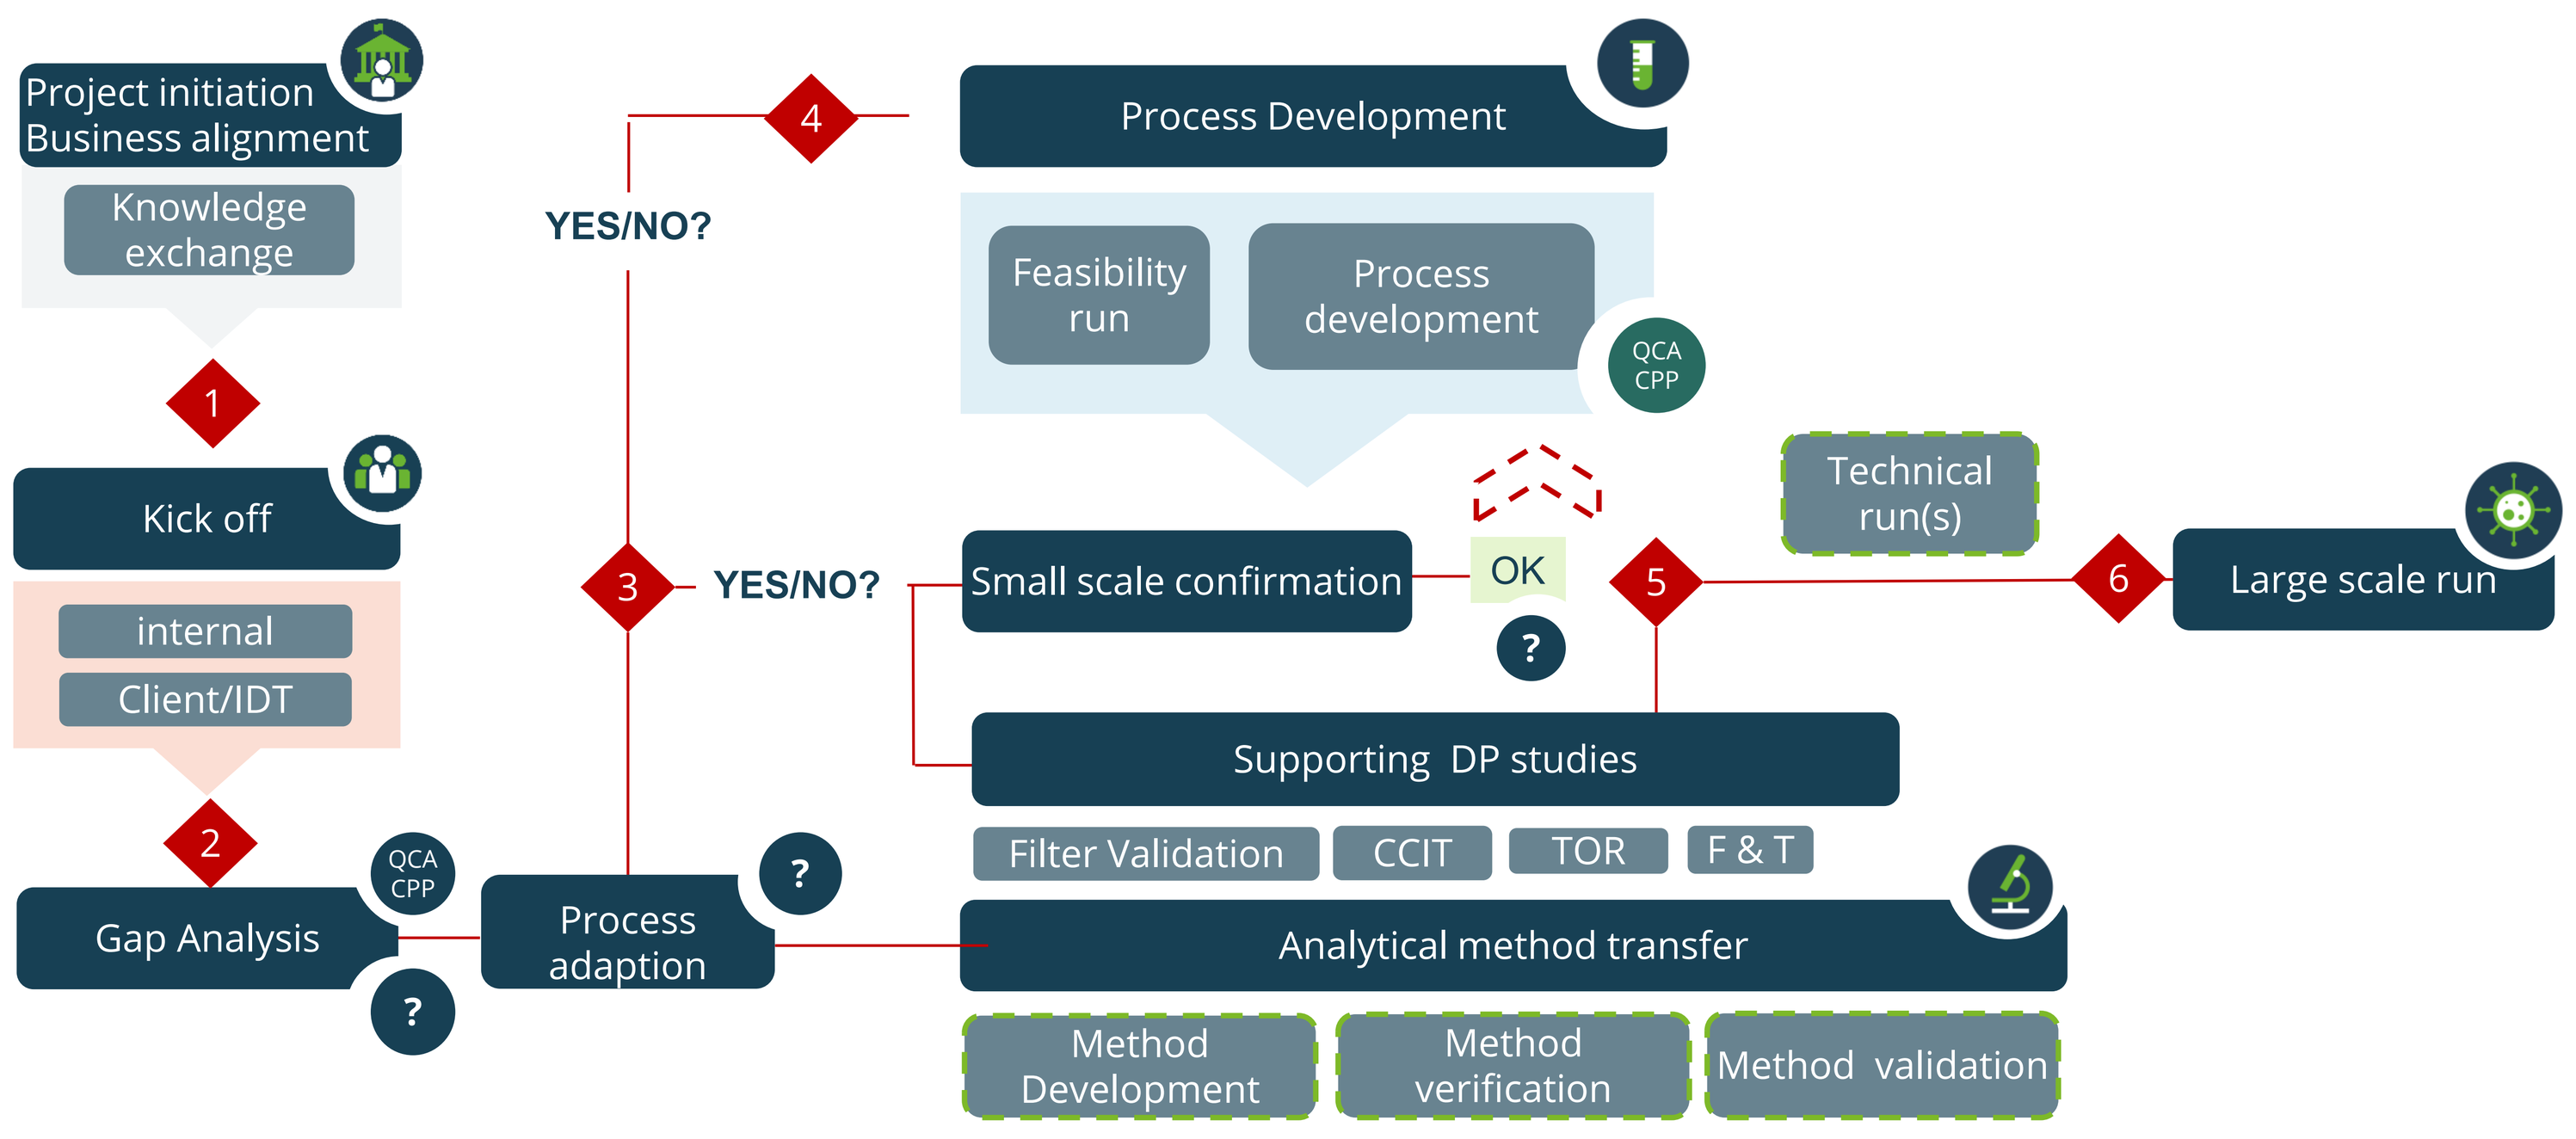
Project initiation
Business alignment
Knowledge exchange
1
Kick off
internal
Client/IDT
2
Gap Analysis
QCA CPP
?
Process adaption
?
3
YES/NO?
4
YES/NO?
Process Development
Feasibility run
Process development
QCA CPP
Small scale confirmation
OK
?
5
Technical run(s)
6
Large scale run
Supporting DP studies
Filter Validation
CCIT
TOR
F & T
Analytical method transfer
Method Development
Method verification
Method validation

Conclusion

- IDT is ready to implement/processes on iCELLis500 technology.
- IDTs flexible process transfer concept is able to implement manufacturing process on every stage of product lifecycle for DS & DP.
- At IDT automated process equipment's for all DSP-MUOs are available.
- Implementation of new equipment possible according to process needs and costumers' requirements.

4. AAV Project

Project starting point:

- GMP process performed at scale for CTM I&II at other CDMOs
- AAV & HEK293 cell line provided by client

Project Target:

- DS & DP manufacturing of CTM phase III with comparable BDS titer
- Implementation of new vial format at established filling line
- Process adaption for GMP compliant CTM Phase III & commercial manufacturing (including PPQ preparation)

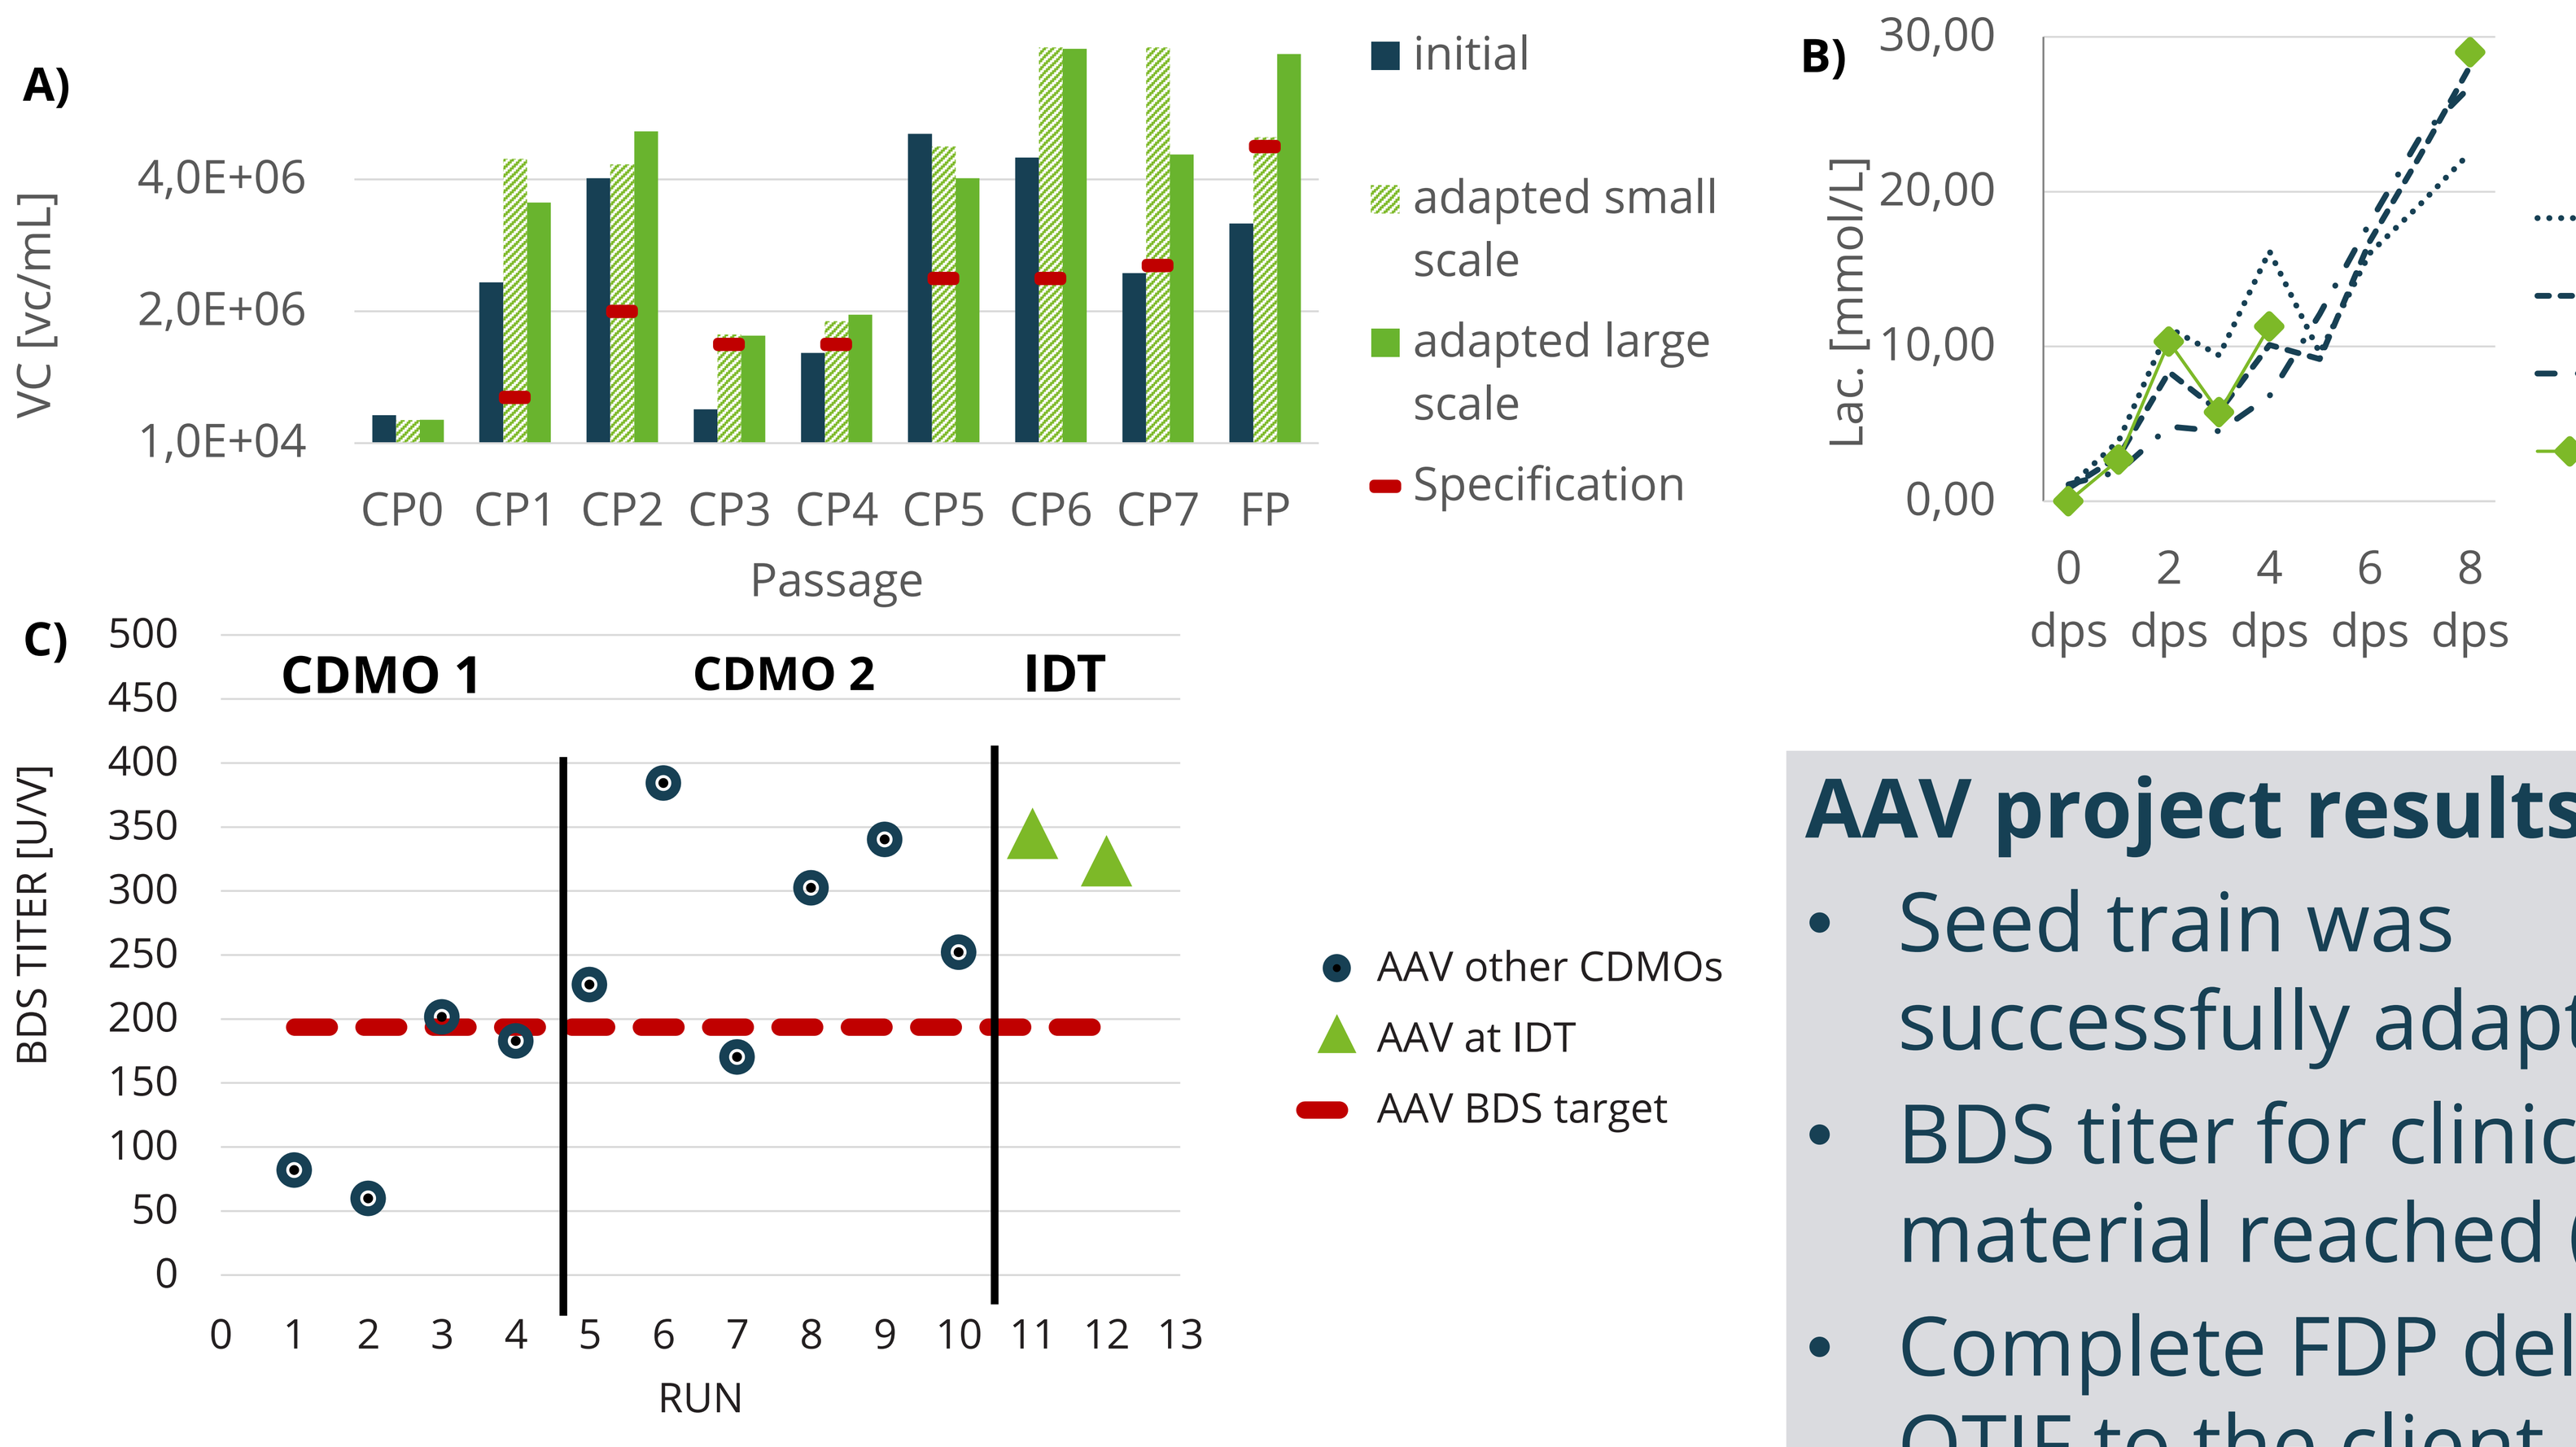
A)
VC [vc/mL]
4,0E+06
2,0E+06
1,0E+04
CP0 CP1 CP2 CP3 CP4 CP5 CP6 CP7 FP
Passage
initial
adapted small scale
adapted large scale
Specification
B)
Lac. [mmol/L]
30,00
20,00
10,00
0,00
0 dps 2 dps 4 dps 6 dps 8 dps
oCDMO R1
oCDMO R2
oCDMO R3
IDT
C)
BDS TITER [U/V]
CDMO 1
CDMO 2
IDT
500 450 400 350 300 250 200 150 100 50 0
0 1 2 3 4 5 6 7 8 9 10 11 12 13
RUN
AAV other CDMOs
AAV at IDT
AAV BDS target

Fig.4: Results of A) adapted seed train, B) metabolic parameters during iCELLis cultivation and C) final BDS titer

AAV project results

- Seed train was successfully adapted
- BDS titer for clinical trail material reached (+75%)
- Complete FDP delivered OTIF to the client

5. Oncolytic Virus Project

Project starting point:

- Lab scale process was developed by customer
- Oncolytic Virus → MSV, WSV established at IDT Biologika
- Adherent human kidney cell line → MCB & WCB established at IDT

Project Target:

- >10x process scale up (final BDS volume) through substitution of CS40 technologies with iCELLis500+ with comparable BDS titer (>1.0E+09)
- **Process development** for GMP compliant CTM Phase III and commercial manufacturing (including PPQ preparation) → substitution of density centrifugation with automated TFF and Chromatography

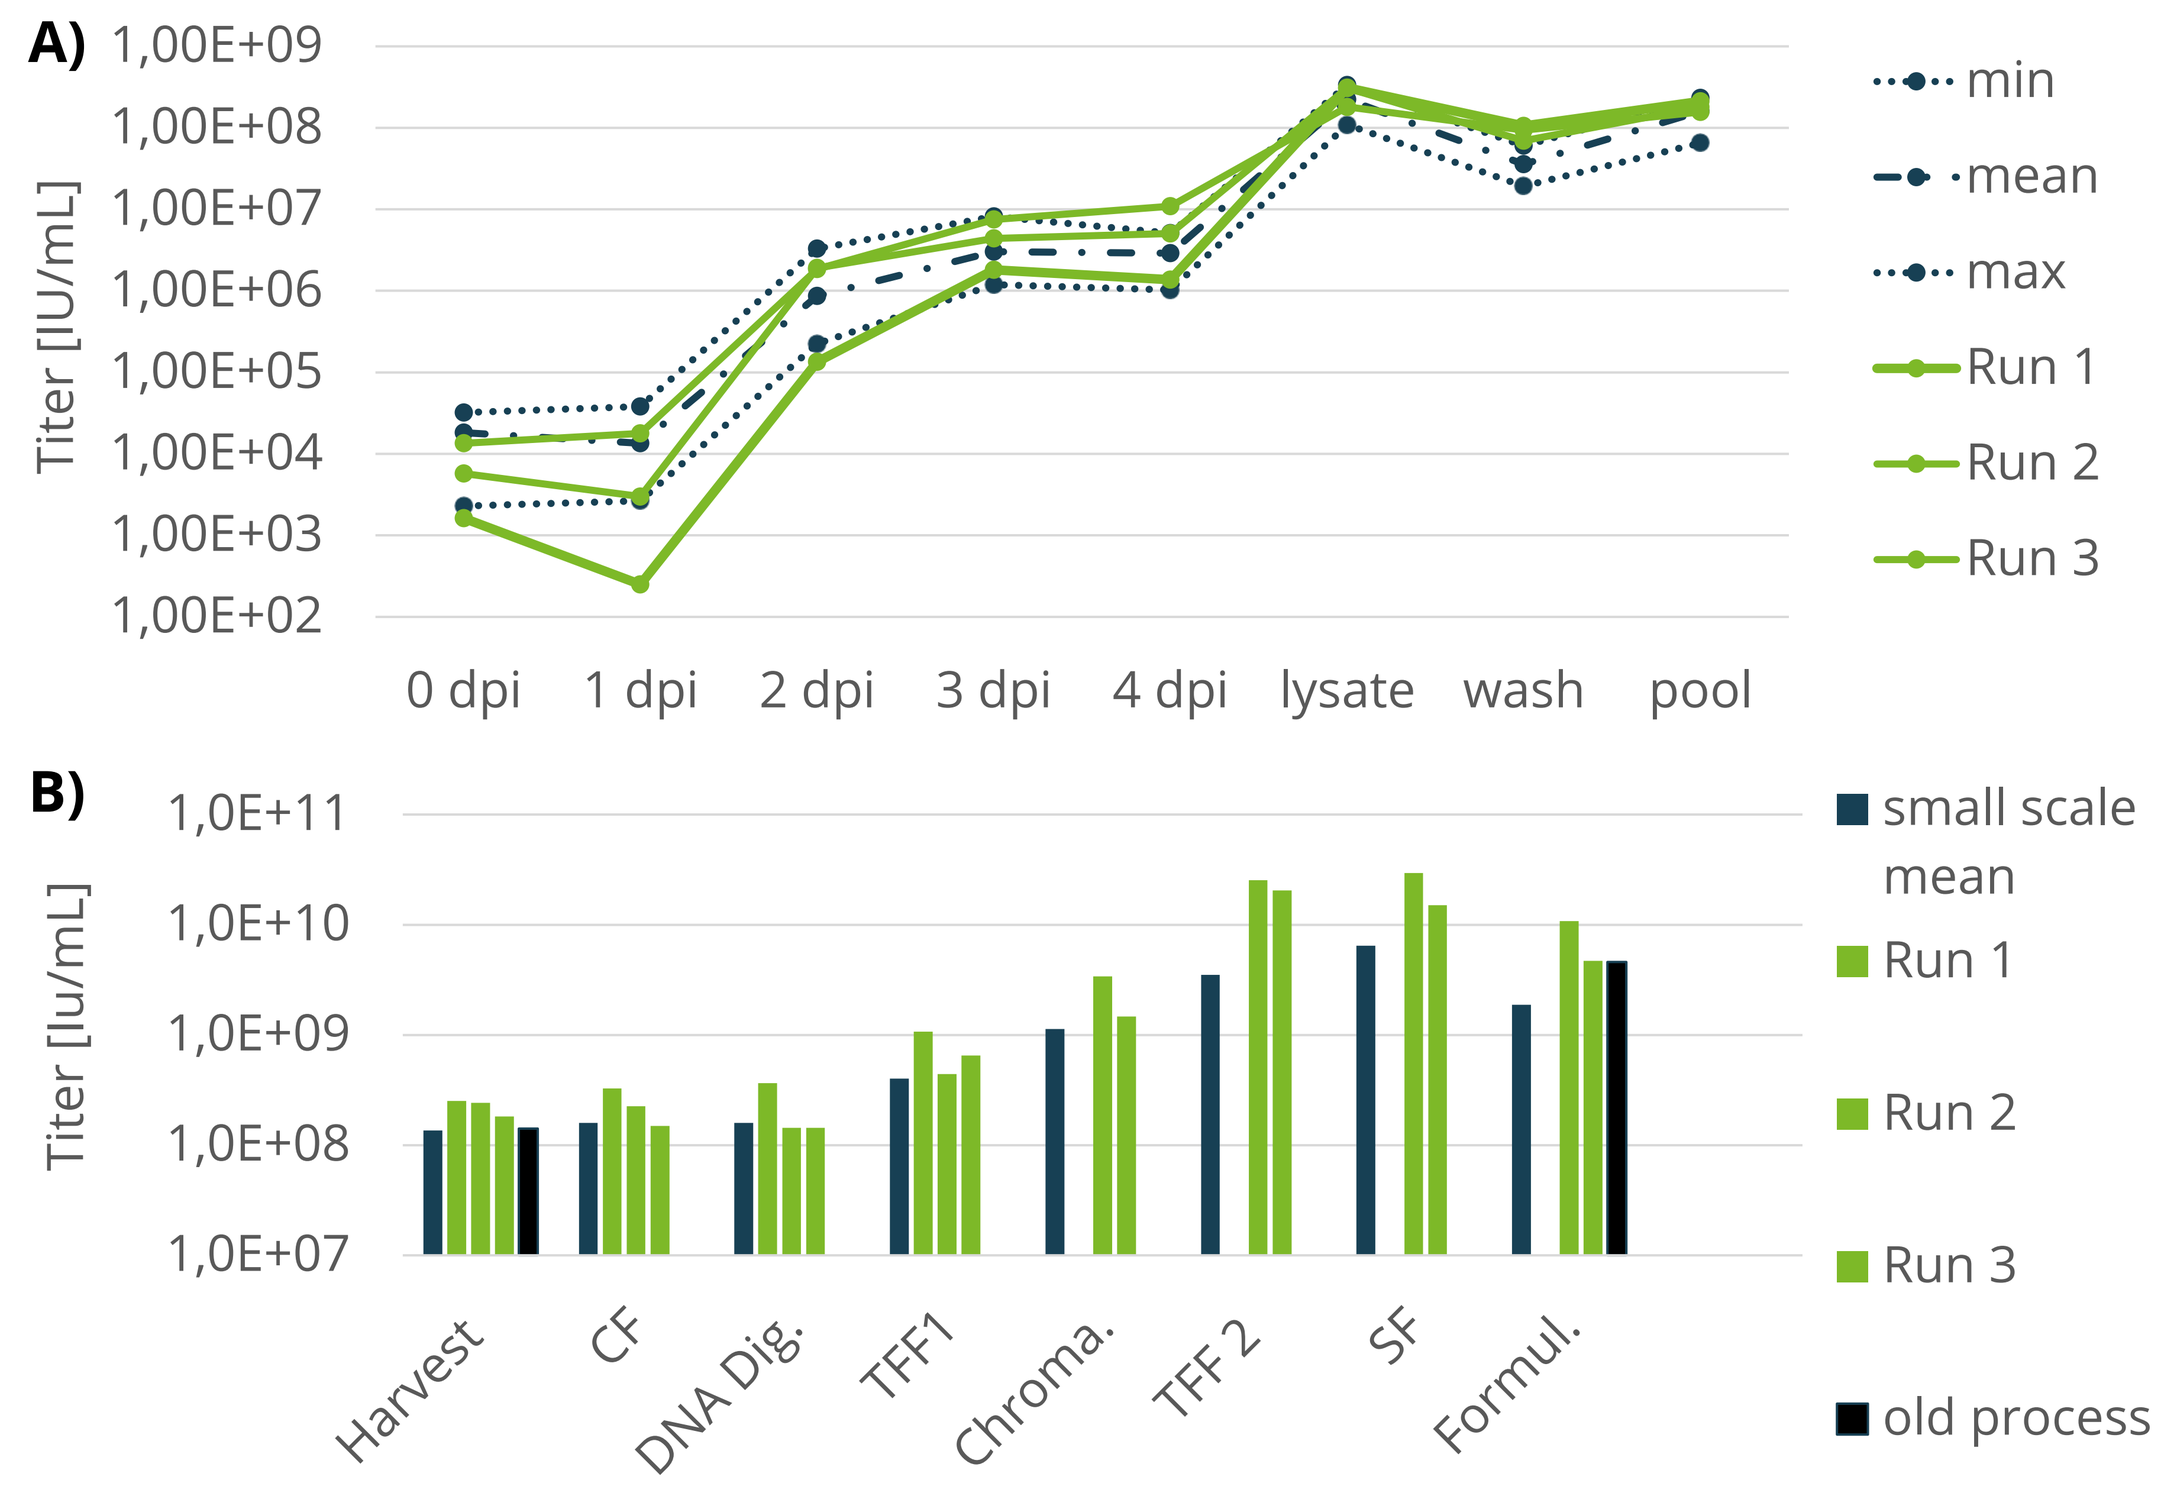
A)
Titer [IU/mL]
1,00E+09
1,00E+08
1,00E+07
1,00E+06
1,00E+05
1,00E+04
1,00E+03
1,00E+02
0 dpi 1 dpi 2 dpi 3 dpi 4 dpi lysate wash pool
min
mean
max
Run 1
Run 2
Run 3
B)
Titer [Iu/mL]
1,0E+11
1,0E+10
1,0E+09
1,0E+08
1,0E+07
Harvest CF DNA Dig. TFF1 Chroma. TFF 2 SF Formul.
small scale mean
Run 1
Run 2
Run 3
old process

Fig.5: Titer results of A) USP and B) DSP process.
Run 1: 1st. Scale up was only performend up to TFF1

Oncolytic Virus Project results

- Successful process development & scale up
- Successful implementation of simplified, scalable closed state of the art USP & DSP process with reduced contamination risk
- Complete FDP delivered OTIF to the client

Contact

Dr. Martin Pech, Senior Manager MSAT & HoM, *Martin.Pech@idt-biologika.de*

www.idt-biologika.com